Synergy Data Analytics: Novel Synergy Score Modeling Analysis Pipeline for Drug-Drug Interactions

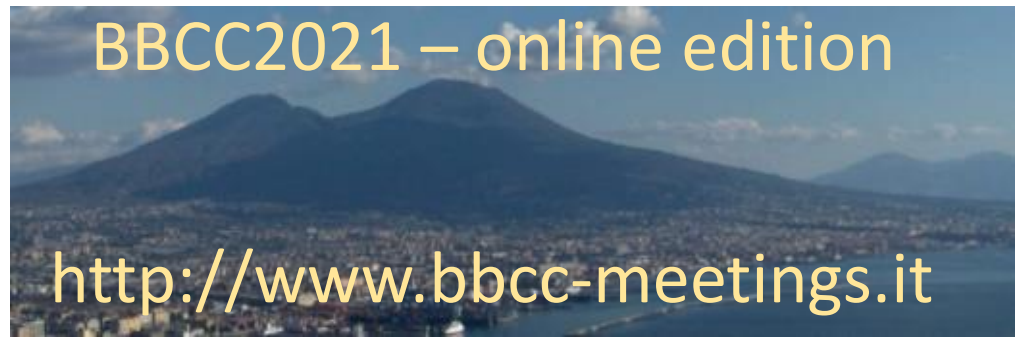

BBCC2021 – online edition
http://www.bbcc-meetings.it

Shiquan Wu[1], Nisebita Sahu[2], Stephen Wong[1], Henry Dewhurst[3], Leo Wang-Kit Cheung[1]

[1]*Bioinformatics Group, GRC, AbbVie Inc., South San Francisco, CA, USA;*
[2]*Oncology Discovery Group, AbbVie Inc., South San Francisco, CA, USA;*
[3]*Bioinformatics Data Engineering Group, GRC, AbbVie Inc., South San Francisco, CA, USA.*

Abstract

Synergy Data Analytics pipeline introduced in this poster is developed with user friendly web-interfaces for synergy score analyses on drug-drug interactions. The pipeline provides a platform to automatically process and analyze drug-drug interaction experimental raw signal data for investigating either pairwise drugs or more than two drugs together with customized graphs and tables. It consists of five major features:

(1) Normalize raw signal data for drug-drug interactions into Inhibition/control percentages through flexible input formats in Excel, CSV, tab-delimited TXT, and RData for various designed plates/wells;
(2) Fit groups of dose-response data by four-parameter log-logistic curves, which achieve better performance than methods that other similar software packages employed;
(3) Compute synergy scores for drug-drug interactions based on various scoring models, e.g. Bliss, Highest Single Agent, and Loewe;
(4) Create various customized score graphs and tables with user-friendly interactive mouse-over information display of metadata of any data points.
(5) This pipeline not only allows investigation of pairwise drug-drug interactions when we have more than 2 drugs to be investigated but also allows investigation of synergy of 3 or more drugs simultaneously for large collections of drug combinations whereas other available software packages may only allow pairwise or small collections of drug-drug interactions for control percentage data.

Introduction

Optimal combinations of drug dosages should be identified to achieve best treatment efficacy when multiple drugs are applied to treatments. This is a critical step for various therapies. A series of software, pipelines or R packages were available for drug synergy analyses, e.g. Combenefit[1], CSS[2] SynergyFinder[3], and mixlow[4].

However, each software or pipeline has disadvantages, e.g. inflexible input formats, visualization problems, lack of or limited customizable analyses, and the numbers of drugs that can be dealt with. More efficient analytics pipeline is greatly needed.

We developed a novel web-based synergy score analysis pipeline, Synergy Data Analytics, to address limitations and disadvantages of publicly available software tools with flexible and lots of customizable analyses.

R Shiny, web-interface and cloud-based analytics are applied to implement the web-based Synergy Data Analytics. The pipeline customizes synergy data analyses for drug-dug interactions from the raw signals produced by biological experiments with enhanced functionalities. It deals with 2 drugs (pairwise combinations) or more than two drugs simultaneously with both internal and Combenefit input formats.

Method

Our Synergy Data Analytics Pipeline is designed to include four major parts: (1) Taking flexible inputs for raw signal data; (2) Normalizing raw signals into control percentages; (3) fitting data by a dose-response four parameter log-logistic curve for each single drug; (4) Analyzing synergy scores by various models, including: Bliss, HSA (Highest Single Agent), and Loewe, followed by creating various types of customized score tables and figures with user-friendly interactive mouse-over information display of metadata of any data points.

Synergy Data Analytics: Mathematical and Statistical Models

Step 1: Normalization (Convert raw response signals into percentages)
Normalized Percentage = (1-Raw Response signal/Reference)x100
Option: Normalized Percentage = 0 if < 0 (i.e. Focus on: Control percentage ONLY)

Step 2: Synergy Score (Compute Prediction Efficacy by 3 types of models)
(1) Bliss: Effect(A+B) = Effect(A) + Effect(B) - Effect(A*B)
(2) HSA: Effect(A+B) = max(Effect(A), Effect(B))
(3) Loewe: Effect(A+B) = f(Effect(A), Effect(B)), same score for all (d_A,d_B) with $d_A/D_{EA} + d_B/D_{EB} = 1$, where D_{EA} & D_{EA} are dosages for Effect(A) & Effect(B).
Synergy Score = Normalization - Prediction Efficacy
In addition, data may be adjusted/corrected by Dose-Response curve fitting.

Web-Interface

Synergy Data Analytics pipeline provides friendly interface for data input and various parameter fields for synergy data analyses from raw signal data to a series of customizable outputs with graphs and tables. It can be set as an automated data analytics pipeline for drug synergy analyses.

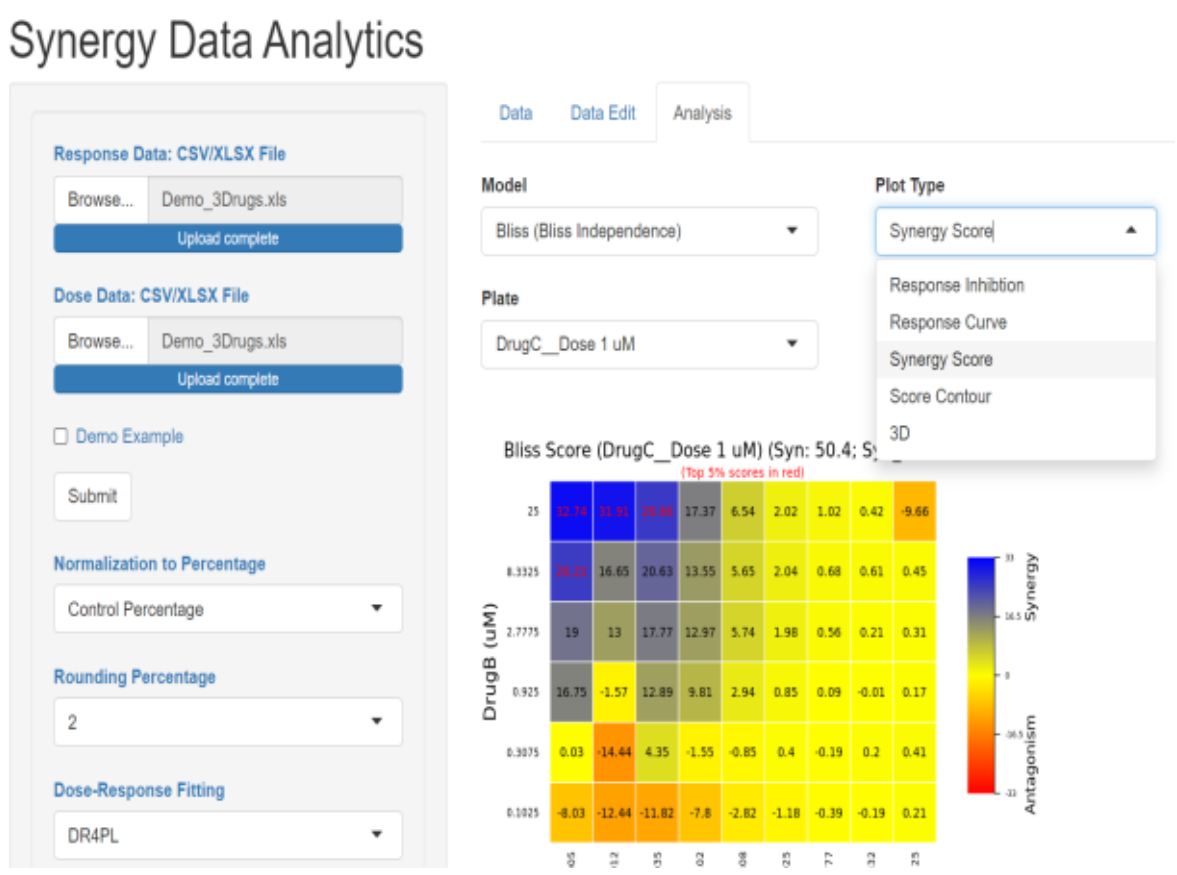

Synergy Data Analytics
Response Data: CSV/XLSX File
Browse... Demo_3Drugs.xls
Upload complete
Dose Data: CSV/XLSX File
Browse... Demo_3Drugs.xls
Upload complete
Demo Example
Submit
Normalization to Percentage
Control Percentage
Rounding Percentage
2
Dose-Response Fitting
DR4PL
Data
Data Edit
Analysis
Model
Bliss (Bliss Independence)
Plot Type
Synergy Score
Response Inhibtion
Response Curve
Synergy Score
Score Contour
3D
Plate
DrugC__Dose 1 uM
Bliss Score (DrugC__Dose 1 uM) (Syn: 50.4; Sy
(Top 5% scores in red)
DrugB (uM)
Synergy
Antagonism

Input Formats

Synergy Data Analytics Pipeline accepts more flexible formats of input than other similar software[1,4], including: Combenefit formatted data input and our Internal inputs: .xls, .xlsx, csv, and txt with 96x96 or 384x384 wells for 2 or more drugs, see illustrated table below. It accepts both raw and normalized data.

All Inputs can be automatically detected and analyzed.

Dosage	25	8.332	8.332	2.777	2.777	0.925	0.925	0.308	0.308	0.102	0.102	0.035	0.035	0.012	0.012	0.005	0.005	0.000	0.000	0.000
25	0.45	0.57	1.75	2.54	3.11	2.88	6.95	8.53	17.12	18.25	22.15	26.61	34.24	28.03	24.3	30	26.78	21.64	19.27	16.1
25	0.45	0.57	1.75	2.54	3.11	2.88	6.95	8.53	17.12	18.25	22.15	26.61	34.24	28.03	24.3	30	26.78	21.64	19.27	16.1
8.332	78.2	65.2	83.57	72.27	77.75	57.07	125.32	100.52	146.63	114.02	158.43	126.17	143.69	132.78	133.63	120.46	127.58	113.06	129.73	96.17
8.332	74.98	70.35	81.25	68.48	76.05	52.6	110.12	96.34	132.61	110.01	137.7	120.46	148.6	120.63	122.84	116.51	119.28	88.43	115.38	98.03
2.777	106	100.91	129	117.75	130.35	128.04	166.97	184.82	177.98	171.54	201.55	199.06	182.56	166.34	156.85	145.61	188.49	161.2	138.49	122.84
2.777	102.21	106.85	123.97	108.15	113.46	103.4	182.5	155.04	177.53	156.63	184.76	174.37	162.28	136.62	152.95	138.26	165.61	136.17	136.57	127.19
0.925	94.59	103.8	119.33	116.74	154.31	115.89	173.86	167.02	194.77	166.01	172.45	164.65	179.4	142.56	176.46	163.52	163.07	144.53	165.16	137.53
0.925	95.66	90.86	105.89	105.66	126.23	106.79	157.08	155.21	174.88	154.93	165.67	152.95	165.89	141.6	165.89	147.02	153.35	142.44	154.88	134.42
0.308	109.79	100.91	110.41	88.03	143.91	138.6	166.01	159.11	180.81	159.34	179.17	159.11	161.2	146.96	152.9	150.58	153.86	132.22	143.46	129.17
0.308	112.78	99.78	98.03	104.59	142.5	128.37	156.01	151.03	157.93	128.32	165.05	145.33	148.26	127.7	149.96	135.49	155.21	121.37	146.4	117.13
0.102	108.99	101.54	127.19	103.68	141.03	127.53	172.05	138.09	152.16	147.47	154.59	150.69	166.29	142.67	153.18	145.1	145.72	140.69	148.89	128.71
0.102	87.01	89.33	93.63	85.55	122.61	130.86	139	135.21	136.4	137.92	132.1	136.06	132.27	141.03	133.35	124.53	128.32	130.64	122.27	126.06
0	101.31	79.84	117.98	117.92	163.97	136.06	180.64	160.13	202.96	166.68	193.81	164.65	160.58	144.25	166.23	138.83	173.13	146.57	134.48	127.19
0	92.38	101.03	109.9	126.17	132.33	132.84	167.36	148.72	171.03	152.33	167.25	144.82	154.2	138.21	150.02	140.35	155.72	139.51	132.78	117.92

Results

Fitting Drug Responses by Dose-Response Curves

Four-parameter log-logistic dose-response curve is applied to fit response data for each drug. Curves are also displayed by HighCharts with interactive mouse-over information for metadata of data points.
Our fitting method is showed by simulations better than those used by similar publicly available software. Fitting data by DRM (dose-response model) based on several types of non-linear functions may have issues in convergence with the functions. Combenefit applies the maximum likelihood approach with a Hill Model to fit data by a three-parameter curve. Mathematically, fitting data with a four-parameter curve always results a better fit than (or at least as good as) with a three-parameter curve for dose-response data. For performance comparison, our Synergy Data Analytics pipeline, Combenefit, and DRM are respectively applied to fit randomly generated simulated drug dose-response data. The following figures at the bottom shows that our pipeline using a four-parameter curve fitting has consistently lower average Residual Sum of Squares (RSS) than Combenefit and DRM in all simulated data sets.

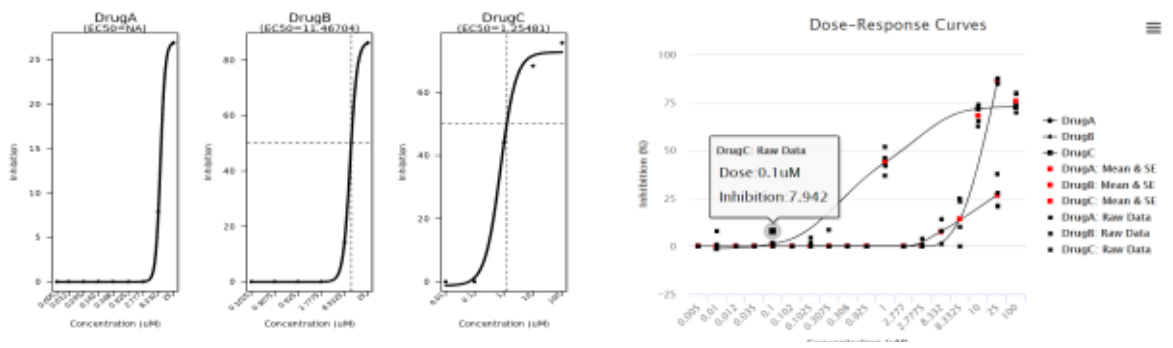

DrugA
DrugB
DrugC
Concentration (uM)
Dose-Response Curves
DrugC: Raw Data
Dose:0.1uM
Inhibition:7.942
Inhibition (%)

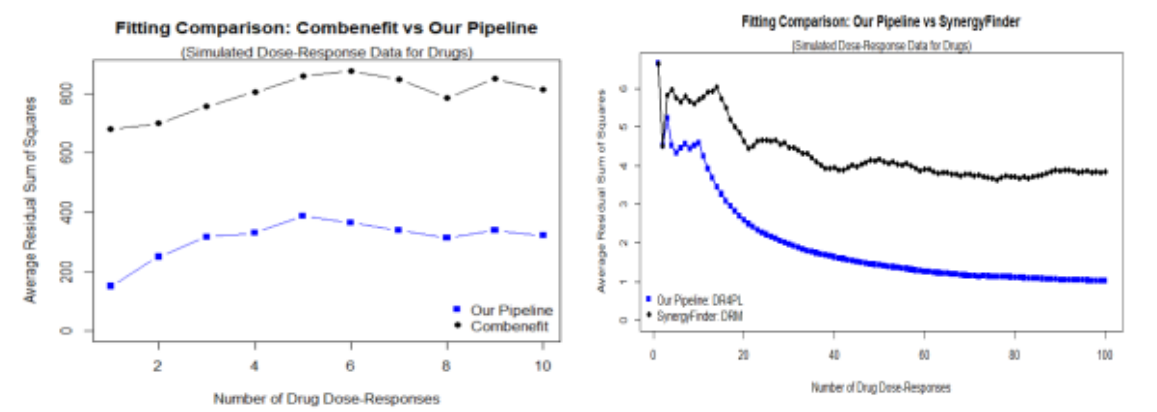

Fitting Comparison: Combenefit vs Our Pipeline
(Simulated Dose-Response Data for Drugs)
Average Residual Sum of Squares
Number of Drug Dose-Responses
Our Pipeline
Combenefit
Fitting Comparison: Our Pipeline vs SynergyFinder
(Simulated Dose-Response Data for Drugs)
Our Pipeline: DR4PL
SynergyFinder: DRM
Number of Drug Dose-Responses

Synergy Scores by Models

Synergy scores are computed by models: Bliss, HSA, and Loewe, respectively. They are visualized by heatmaps, contours, and 3-D displays. All visualizations could be exported for publication easily.

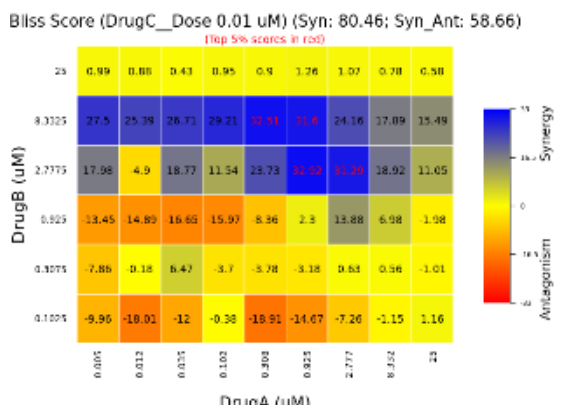

Bliss Score (DrugC__Dose 0.01 uM) (Syn: 80.46; Syn_Ant: 58.66)
DrugB (uM)
DrugA (uM)
Synergy
Antagonism

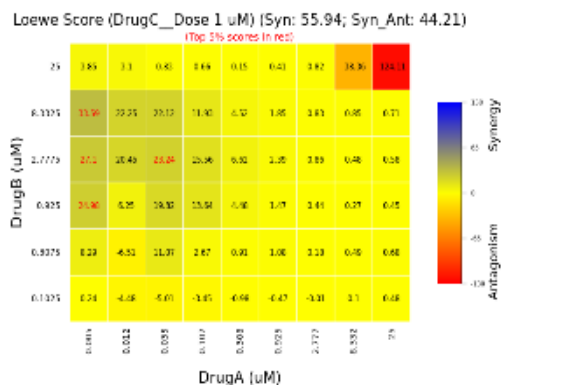

Loewe Score (DrugC__Dose 1 uM) (Syn: 55.94; Syn_Ant: 44.21)
DrugB (uM)
DrugA (uM)
Synergy
Antagonism

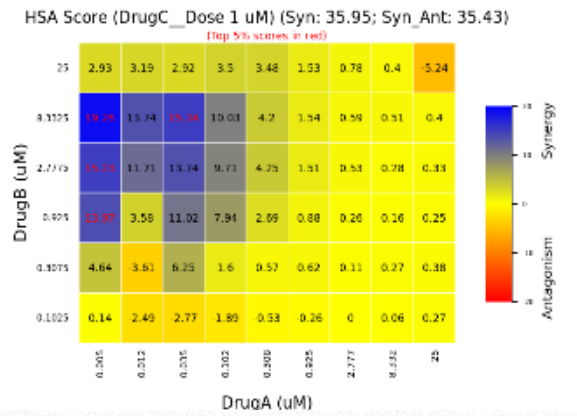

HSA Score (DrugC__Dose 1 uM) (Syn: 35.95; Syn_Ant: 35.43)
DrugB (uM)
DrugA (uM)
Synergy
Antagonism

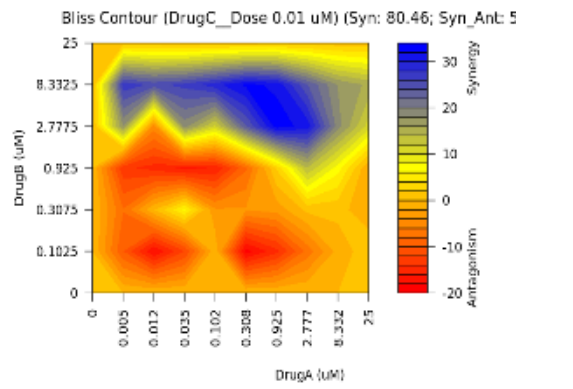

Bliss Contour (DrugC__Dose 0.01 uM) (Syn: 80.46; Syn_Ant: 5
DrugB (uM)
DrugA (uM)
Synergy
Antagonism

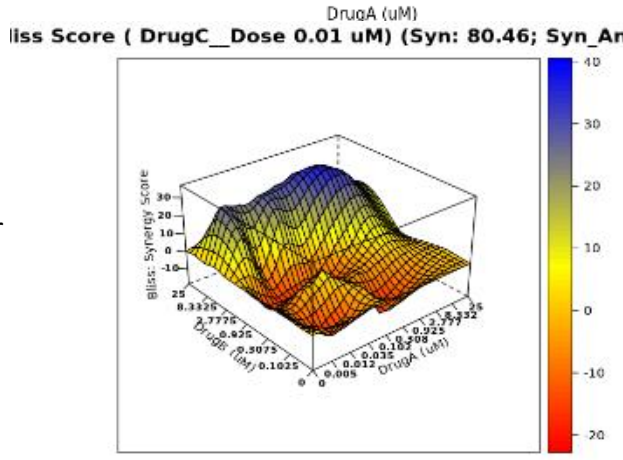

Bliss Score (DrugC__Dose 0.01 uM) (Syn: 80.46; Syn_Ant: 58.6
Bliss: Synergy Score
DrugB (uM)
DrugA (uM)

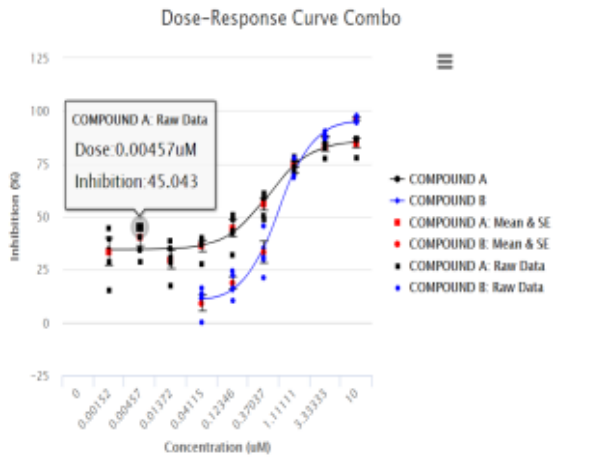

Dose-Response Curve Combo
COMPOUND A: Raw Data
Dose:0.00457uM
Inhibition:45.043
Inhibition (%)
Concentration (uM)
COMPOUND A
COMPOUND B
COMPOUND A: Mean & SE
COMPOUND B: Mean & SE
COMPOUND A: Raw Data
COMPOUND B: Raw Data

Conclusion

Our Synergy Data Analytics pipeline is a convenient customizable web-based automation of synergy data analyses for drug-drug interaction with a user-friendly interface, more options for input files, stronger data processing, and better data fitting and analytics, and high-quality graphical visualizations. Our data fitting has better performance than other similar software packages/pipelines such as Combenefit and SynergyFinder. Our Synergy Data Analytics Pipeline will be made available at GitHub with our full manuscript publication.

References

Di Veroli,G.Y. et al. Combenefit: an interactive platform for the analysis and visualization of drug combinations. Bioinformatics. 2016;32,:2866-82868. doi:10.1093/bioinformatics/btw230.

Alina Malyutina, et al. Drug combination sensitivity scoring facilitates the discovery of synergistic and efficacious drug combinations in cancer. PLOS Computational Biology. 2019; 15,: e1006752. doi:10.1371/journal.pcbi.1006752.

Ianevski A. et al. SynergyFinder: a web application for analyzing drug combination dose–response matrix data. Bioinformatics. 2017; 33,:2413–2415, doi: 10.1093/bioinformatics/btx162.

mixlow: An R Package for Assessing Drug Synergism / Antagonism. https://www.jstatsoft.org/article/view/v034i06/v34i06.pdf3.